EFFECT OF E-101 SOLUTION AND ITS OXIDATIVE PRODUCTS ON MICROBIAL ULTRASTUCTURE CHANGES ASSOCIATED WITH MICROBICIDAL ACTION

Indiana University Health

G. A. Denys[1], M.P. Goheen[1], R.C. Allen[2], P. O'Hanley[3], and J. T. Stephens, Jr.[3]

[1]IU Health Pathology Laboratory, Indianapolis, IN, [2]Creighton Univ. Med. Ctr., Omaha, NE, [3]Exoxemis, Inc., Little Rock, AR.

Poster #1453
21st European Congress of Clinical Microbiology and Infectious Diseases and 27th International Congress of Chemotherapy.
Milan, Italy, 7-10 May 2011

Contact: medinfo@exoxemis.com

ABSTRACT

Objective: E-101 Solution (E-101) is a novel cell free myeloperoxidase-mediated antimicrobial developed for topical application directly into surgical wounds. It is composed of 1) porcine myeloperoxidase (pMPO) and glucose oxidase (GO), 2) glucose, 3) sodium chloride, and 4) specific amino acids. Once activated, hydrogen peroxide (H_2O_2) is produced in situ by GO dehydrogenation of glucose and reduction of oxygen. The MPO-catalyzed oxidation of chloride by H_2O_2 generates hypochlorous acid (HOCl). Once generated, HOCl reacts in a diffusion-controlled reaction with a second H_2O_2 molecule to yield singlet oxygen. We evaluated the treatment effects of E-101 and its oxidative products on ultrastucture changes and microbicidal activity against methicillin-resistant *Staphylococcus aureus* (MRSA) and *Escherichia coli*.
Methods: Time kill and transmission electron microscopy (TEM) studies were performed on MRSA ATCC 43300 and *E. coli* ATCC 25922 using proposed therapeutic doses of E-101 (150 and 300 GU/mL) and formulations with pMPO or GO omitted. For TEM, early exponential phase growth were exposed to each formulation and sampled at 0, 30, 60, and 120 min. Bacteria were fixed in 3.0% glutaraldehyde, embedded and thin sectioned for analysis.
Results: E-101 demonstrated rapid (at 30 min) bactericidal activity. At 30 mins morphologic changes were minimal, but at 60 and 120 min dramatic effects on *S. aureus* and *E. coli* morphology were observed characterized as septal deformation. Both formulations of E-101 induced cytoplasmic membrane inclusions (mesosomes) in *S. aureus* indicating a membrane effect. Increase vacuolation of the cytoplasm and cell ghosts were also observed. *E. coli* appeared elongated with no visible septa, highly vacuolated cytoplasm with pleated cell walls compared to controls. When GO was omitted from the formulation no antimicrobial activity or cellular damage was observed. When pMPO was omitted from the formulation, only increased vacuolation due to H_2O_2 was observed at the longer exposure times. Prolonged exposure to high levels of H_2O_2 generated from GO and glucose produced microbicidal activity with minimal cellular damage.
Conclusions: E-101 is a potent myeloperoxidase enzyme system with multiple oxidative mechanisms of action. Ultrastuctural analysis following E-101 treatment showed targeted septum formation in both *S. aureus* and *E. coli*. Induction of mesosomes in *S. aureus* by E-101 is indicative of an effect on the cytoplasmic membrane.

INTRODUCTION AND PURPOSE

E-101 Solution (E-101) is a cell-free coupled enzyme system generating oxidant that acts via multiple mechanisms of action against a broad spectrum of Gram positive and Gram-negative bacteria. It is developed as a microbicide for application directly into wound or surgical incision sites. Hydrogen peroxide (H_2O_2) is produced in situ by glucose oxidase (GO) dehydrogenation of glucose resulting in the two equivalent reduction of oxygen. The acid (H^+) optimum myeloperoxidase-oxidation of chloride ion (the Cl^- of NaCl) by H_2O_2 generates hypochlorous acid (HOCl). Once generated, HOCl (and its conjugate base OCl^-) participates in a diffusion controlled reaction with a second H_2O_2 molecule to yield singlet oxygen ($^1O_2$). Singlet oxygen is a potent electrophilic oxygenating agent capable of reacting with a broad spectrum of electron rich compounds (Figure 1). The mechanism of action of E-101 is enhanced by the binding of pMPO to the surface of target microorganisms.
The microbicidal combustive action of E-101 against target microorganisms is directed to a variety of molecular and enzymatic sites that are essential for metabolism or for the integrity of the microorganism. This study was undertaken to examine the time- and concentration-dependent effects of E-101 and its oxidative products on the ultrastucture of MRSA and *E. coli*.

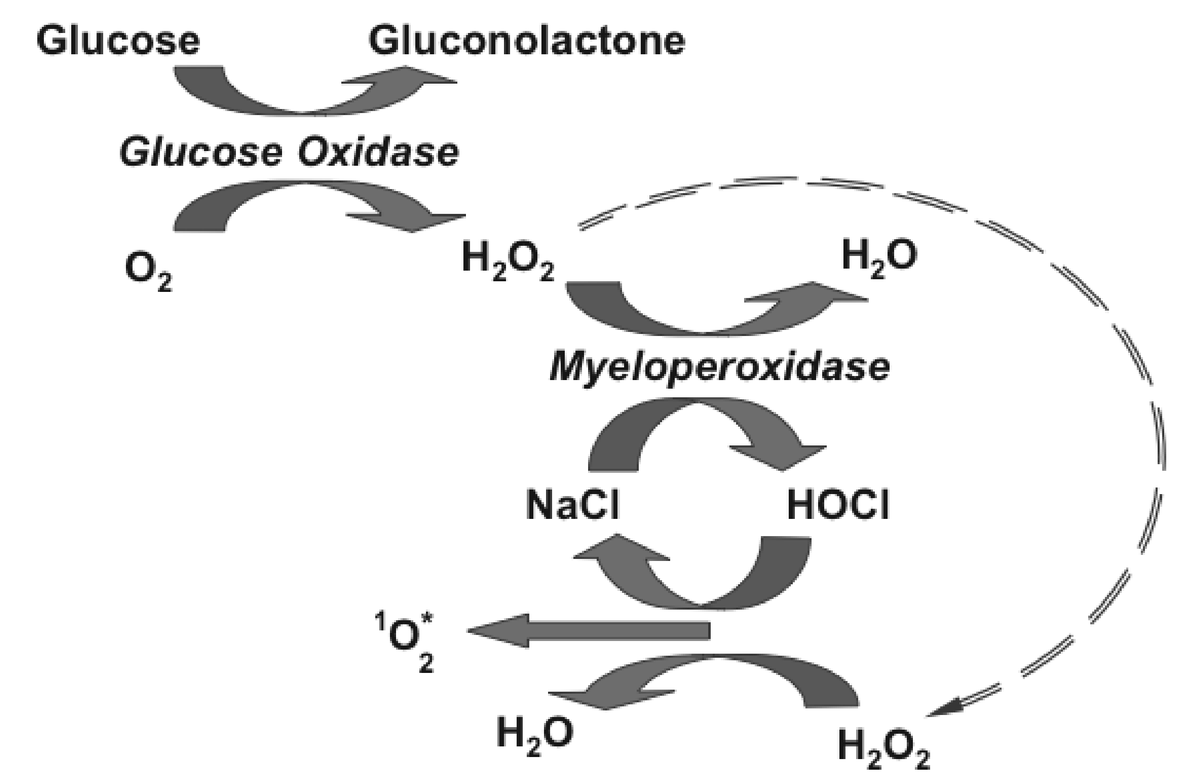
Glucose
Gluconolactone
Glucose Oxidase
O_2
H_2O_2
H_2O
Myeloperoxidase
NaCl
HOCl
$^1O_2^*$
H_2O
H_2O_2

Figure 1. Mechanism of action of E-101 Solution

RESULTS

Figures 2. Time-kill curves of MRSA ATCC 43300 demonstrate rapid bactericidal activity of E-101 (pMPO + GO). High levels of H_2O_2 generated from glucose and GO showed some bactericidal activity but at a much slower rate. Catalase production by *S. aureus* competitively destroys H_2O_2 but was ineffectice in preventing the rapid killing activity of E-101. Formulation without GO (pMPO only) showed no antimicrobial activity.

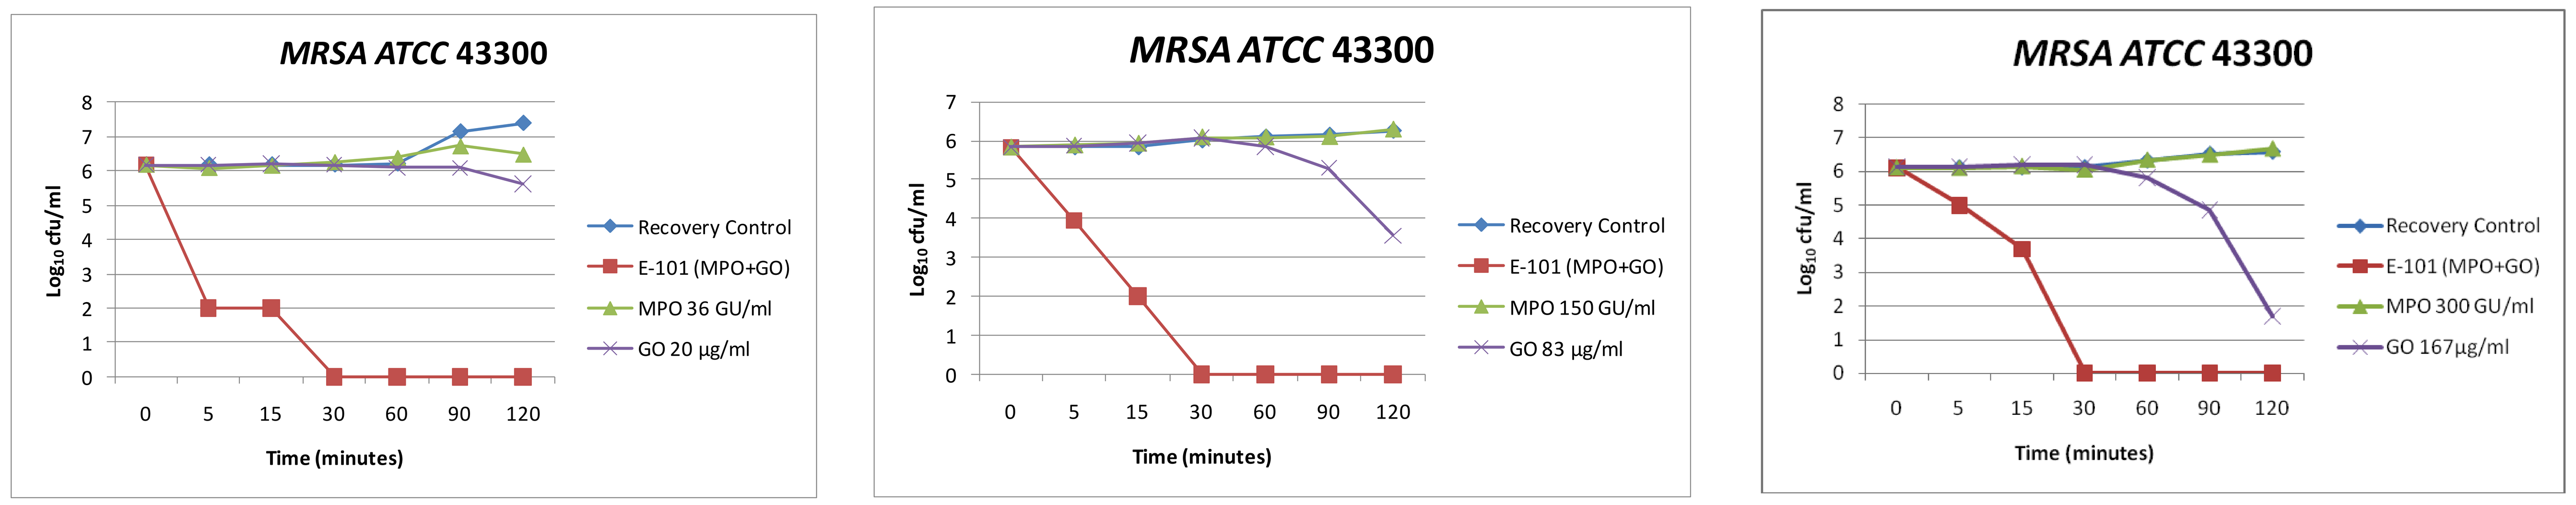
MRSA ATCC 43300
Log_{10} cfu/ml
Time (minutes)
0 5 15 30 60 90 120
Recovery Control
E-101 (MPO+GO)
MPO 36 GU/ml
GO 20 µg/ml
MRSA ATCC 43300
Log_{10} cfu/ml
Time (minutes)
Recovery Control
E-101 (MPO+GO)
MPO 150 GU/ml
GO 83 µg/ml
MRSA ATCC 43300
Log_{10} cfu/ml
Time (minutes)
Recovery Control
E-101 (MPO+GO)
MPO 300 GU/ml
GO 167µg/ml

Figures 3. Time-kill curves of *E. coli* ATCC 25922 demonstrate rapid bactericidal activity of E-101 (pMPO + GO). High levels of H_2O_2 generated from glucose and GO showed bactericidal activity at a much slower rate. Weak catalase production by *E. coli* was inefffective in preventing E-101 killing. Formulation without GO (pMPO only) showed no antimicrobial activity.

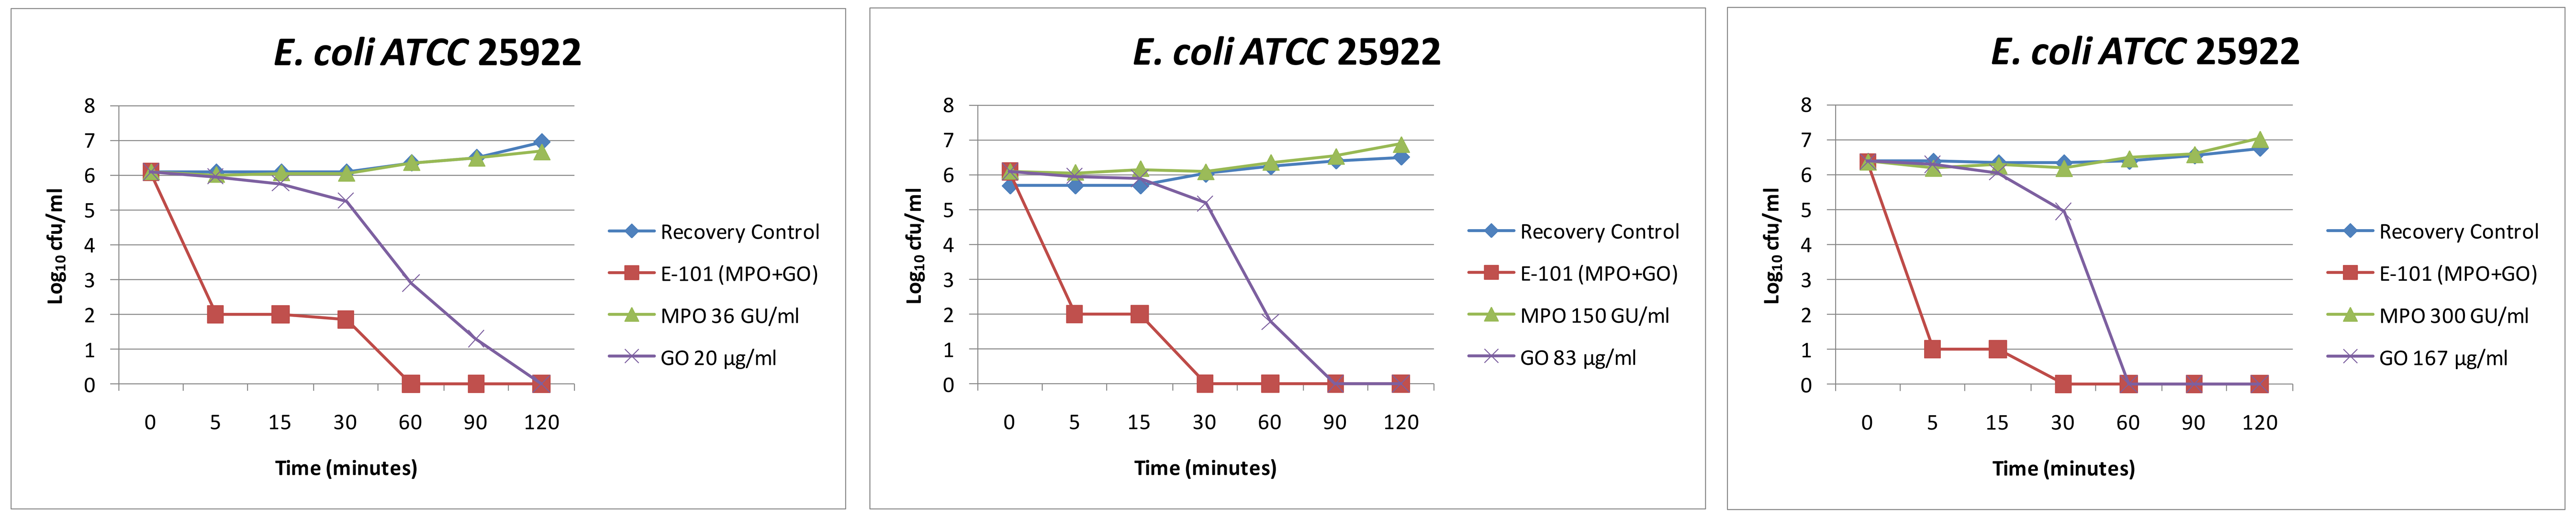
E. coli ATCC 25922
Log_{10} cfu/ml
Time (minutes)
0 5 15 30 60 90 120
Recovery Control
E-101 (MPO+GO)
MPO 36 GU/ml
GO 20 µg/ml
E. coli ATCC 25922
Log_{10} cfu/ml
Time (minutes)
Recovery Control
E-101 (MPO+GO)
MPO 150 GU/ml
GO 83 µg/ml
E. coli ATCC 25922
Log_{10} cfu/ml
Time (minutes)
Recovery Control
E-101 (MPO+GO)
MPO 300 GU/ml
GO 167 µg/ml

Figure 4. E-101 induces substantial time- and concentration-dependent morphological changes in the cytoplasmic membrane of MRSA at 60 and 120 minutes. No apparent ultrastructure changes were observed at 30 minutes and no changes in cultures treated with partial formulation containing GO (H_2O_2 generated) or pMPO only.

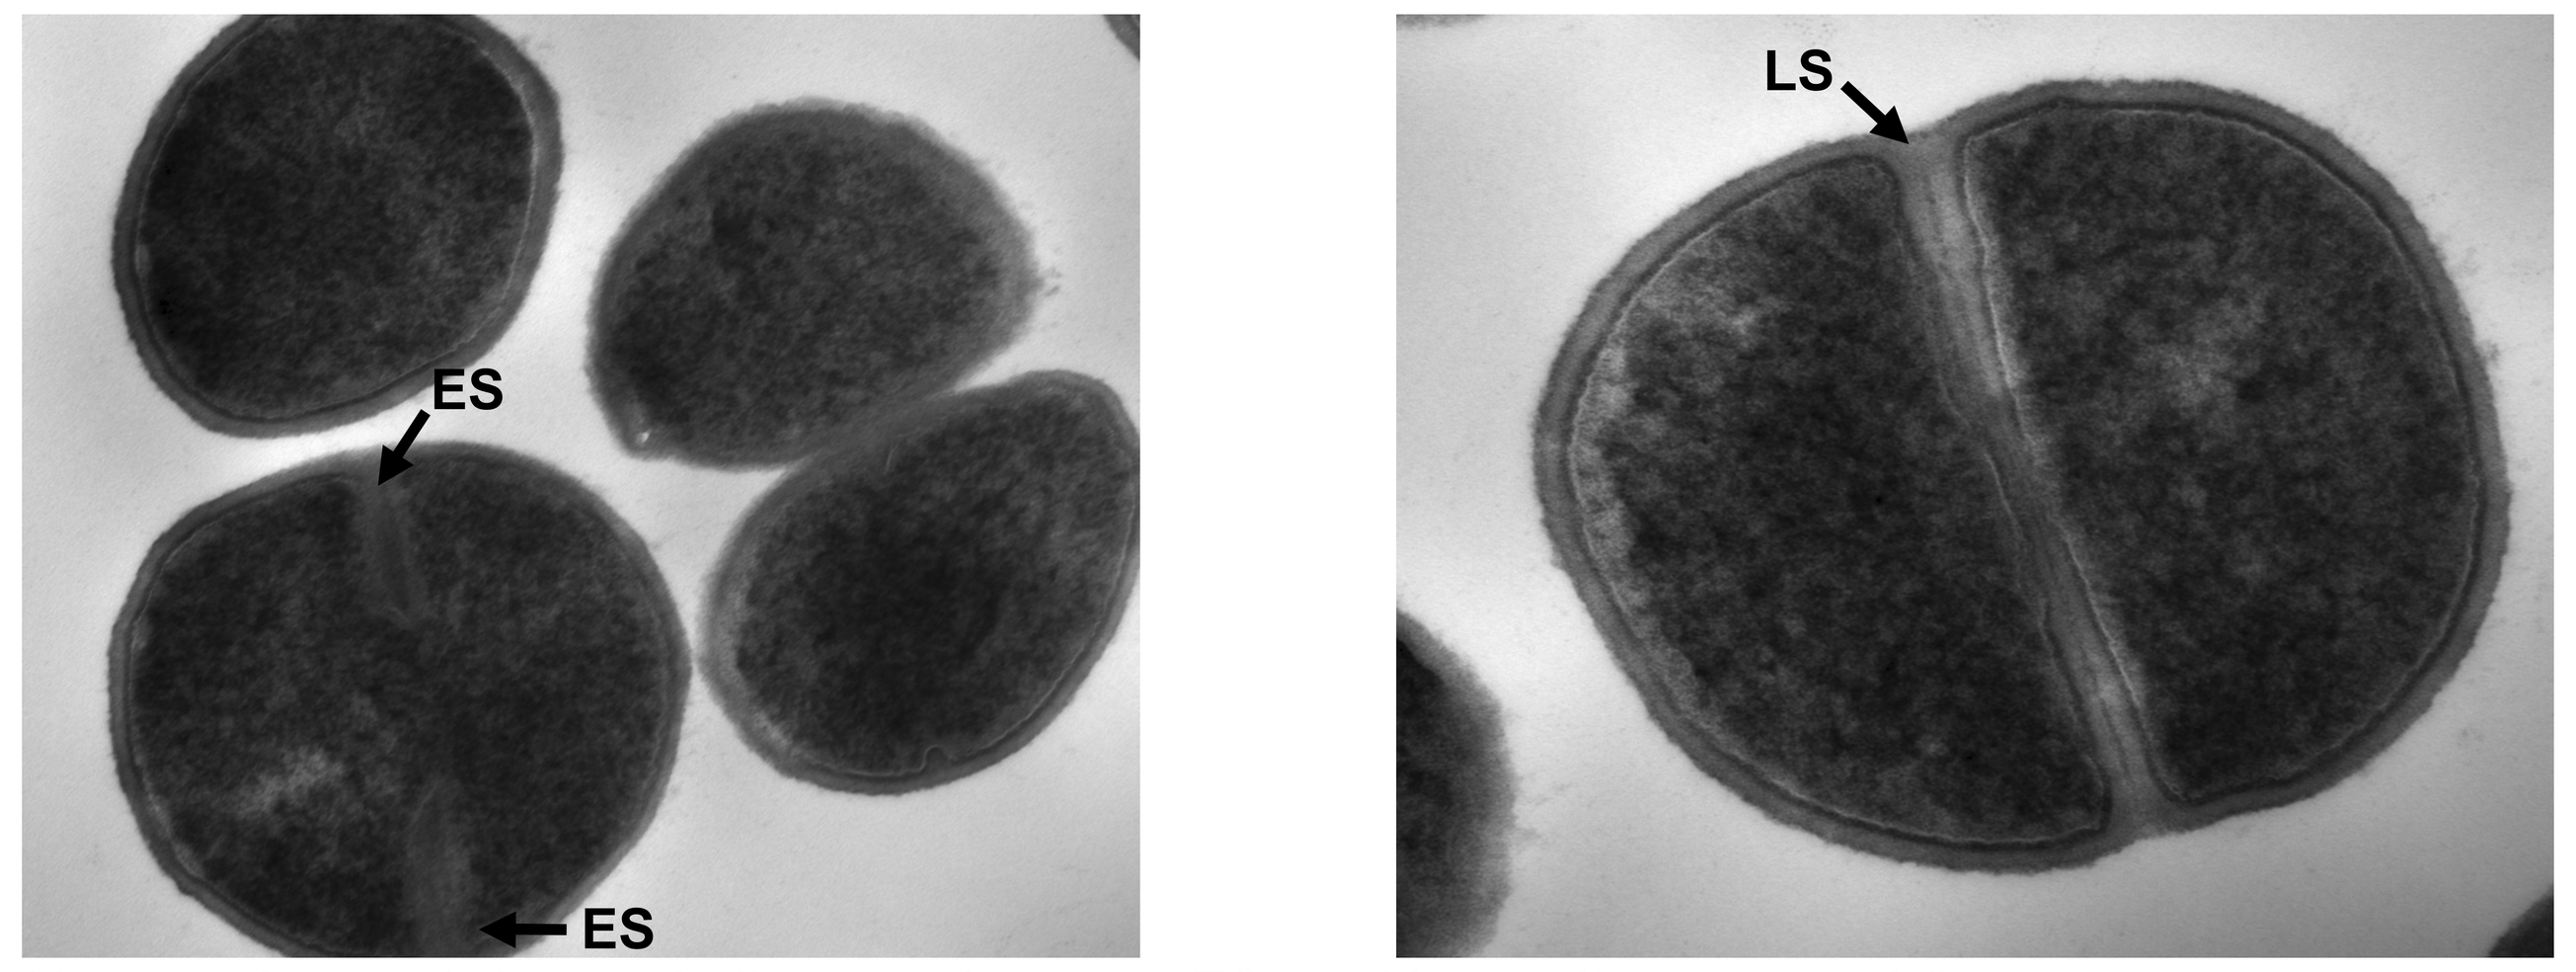
ES
ES
LS

Untreated control. Arrows indicate early septa (ES) and late uniform septa (LS).
Left: 49000x, Right: 68000x

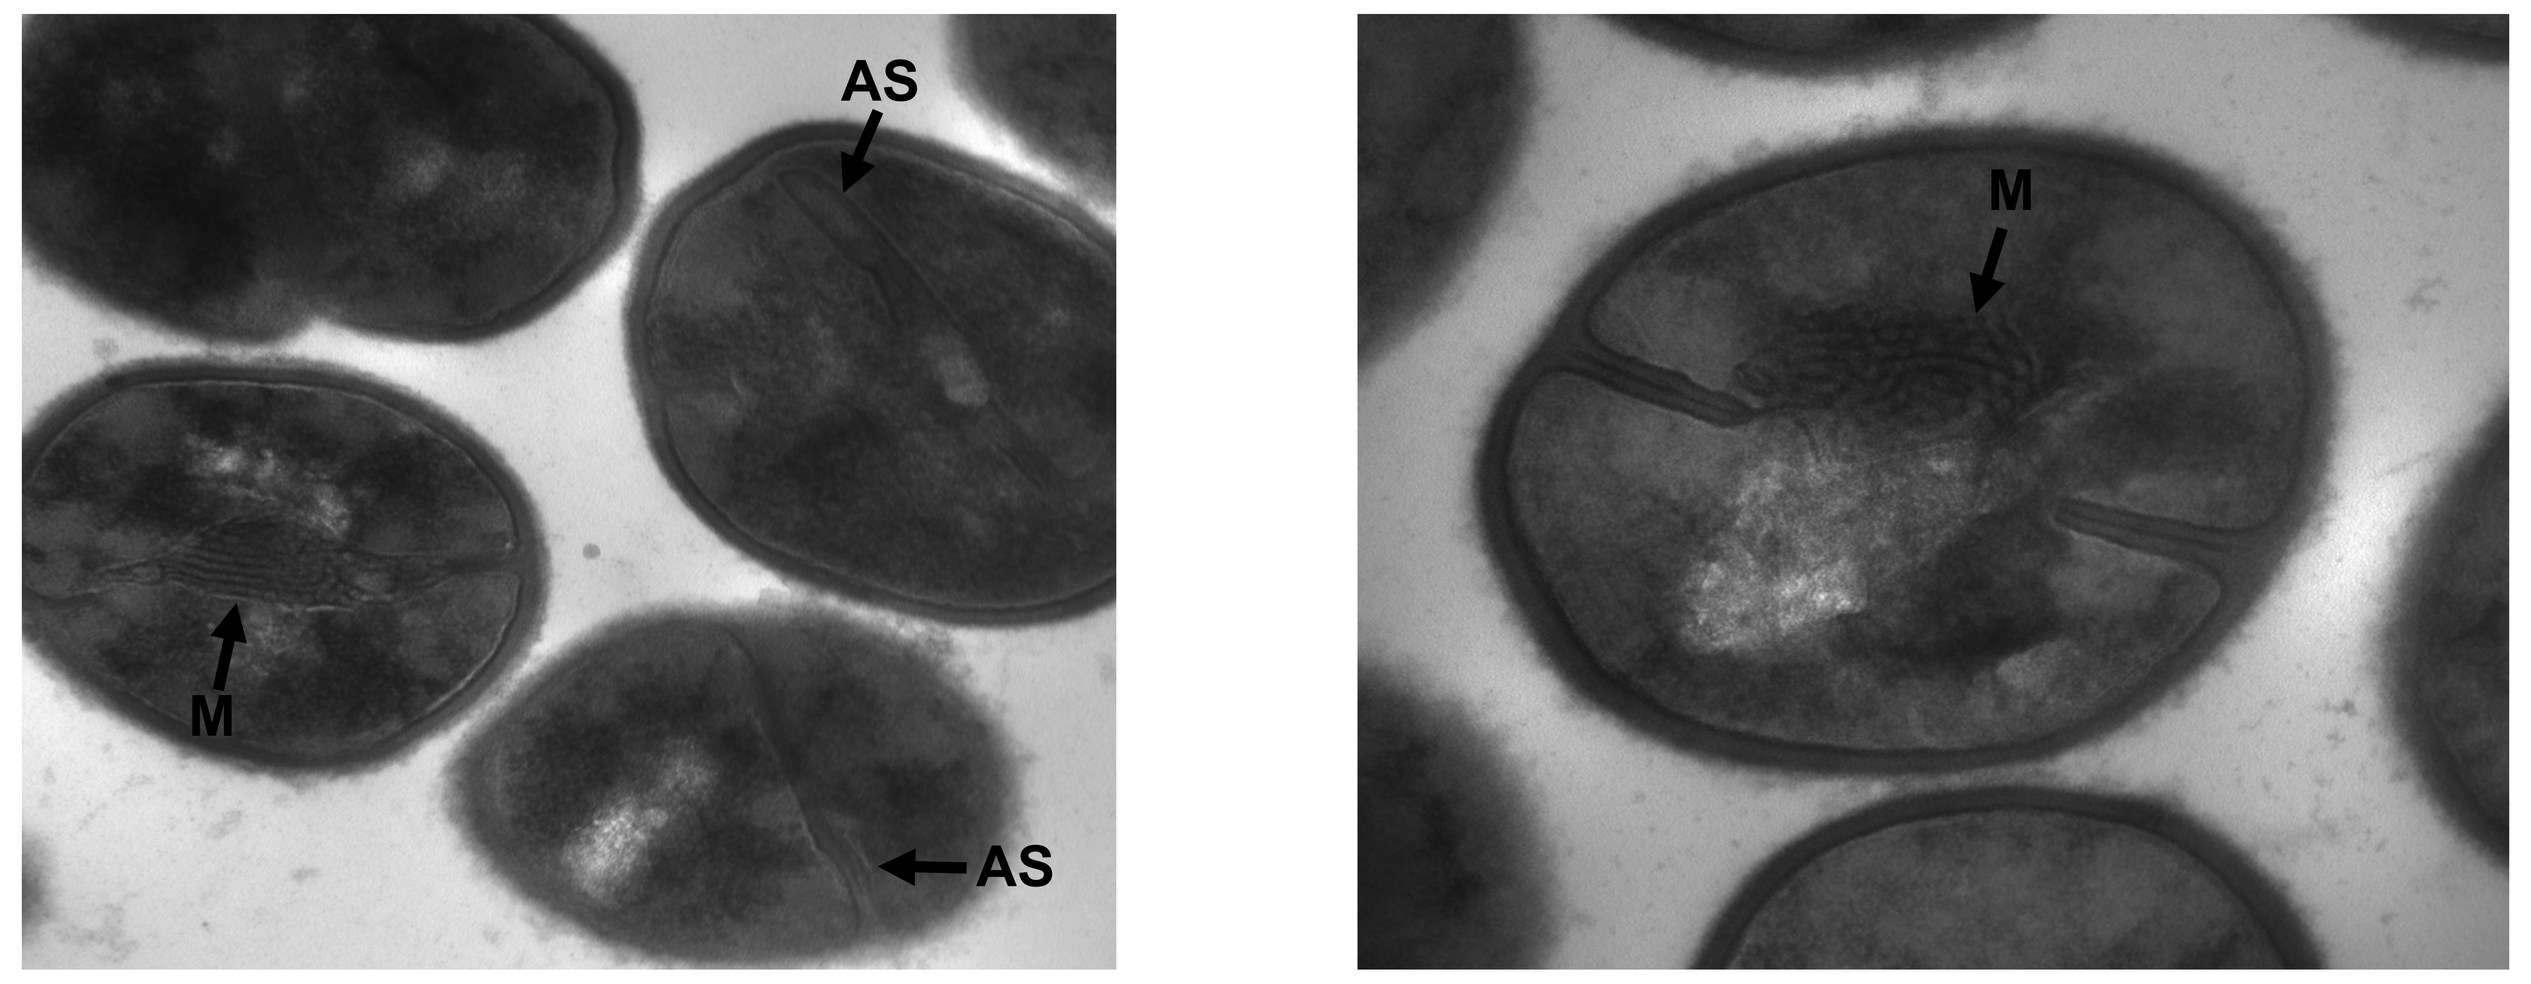
AS
M
AS
M

E-101 causes septal deformation and cytoplasmic membrane (mesosomes). Arrows indicate aberrant septa (AS) and mesosomes (M).
Left: 49000x, Right: 68000x

Figure 5. E-101 induces substantial time- and concentration-dependent morphological changes in septal formation and cytoplasm of *E. coli* at 60 and 120 minutes. Cultures treated with partial formulation containing GO (H_2O_2 generated) or pMPO only demonstrated increase vacuolization after prolonged exposure, but no abnormal cytoplasmic membrane or cell wall changes.

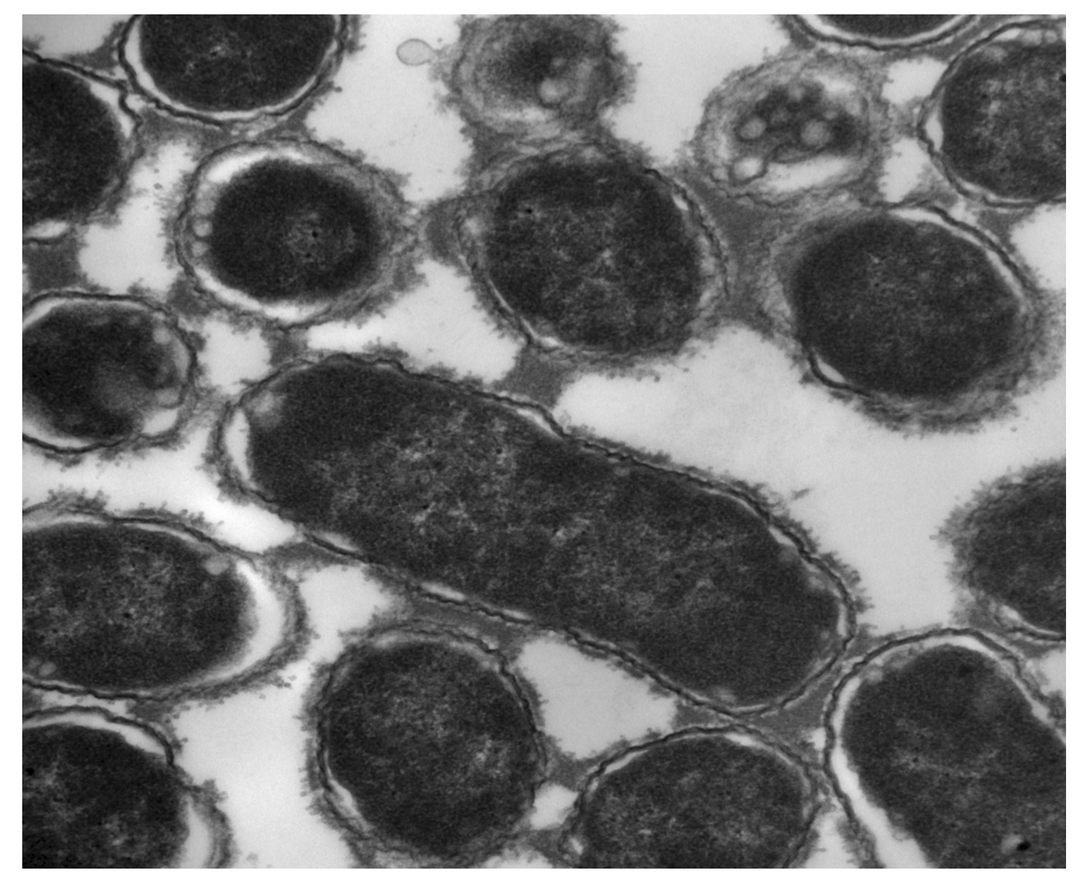

Untreated control. 23000x

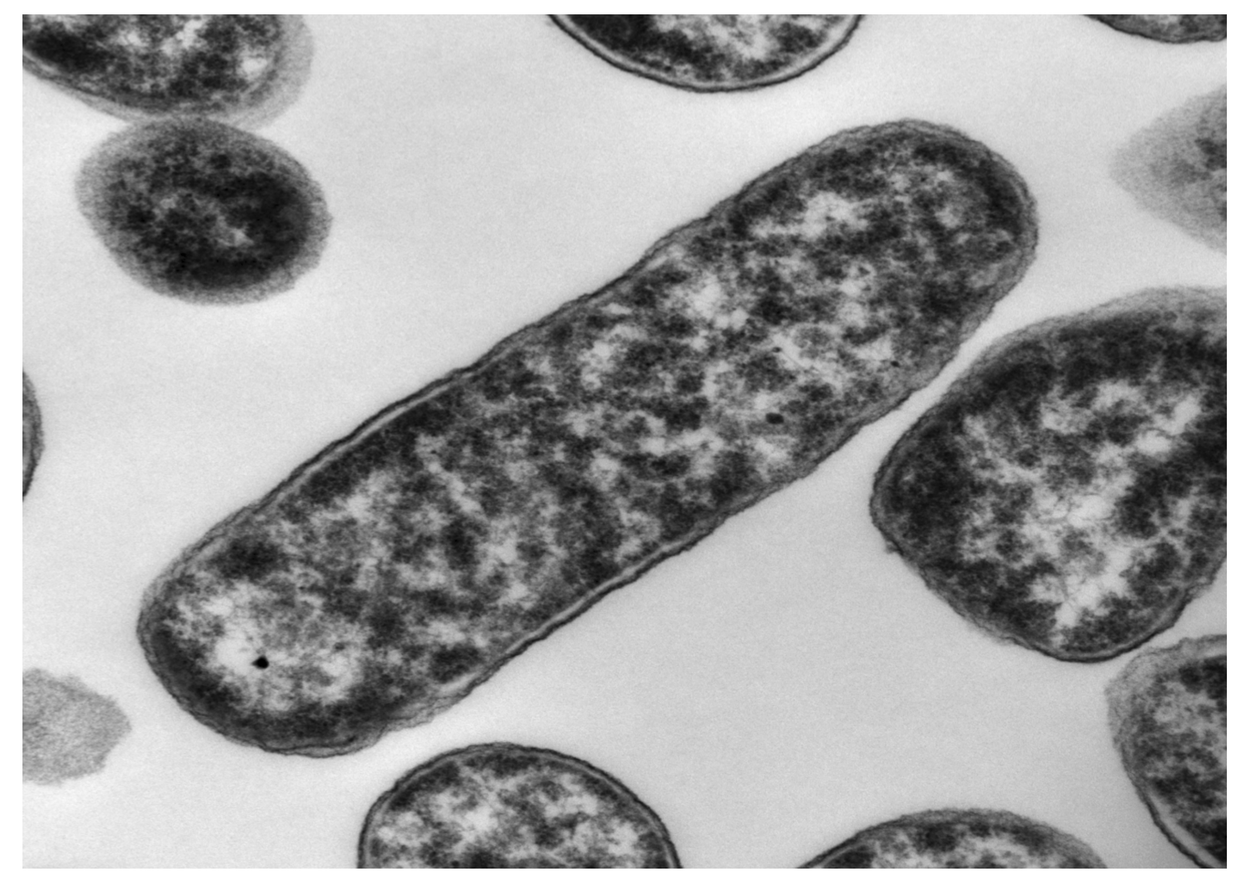

Cultures treated with H_2O_2 generated formulation. 30000x

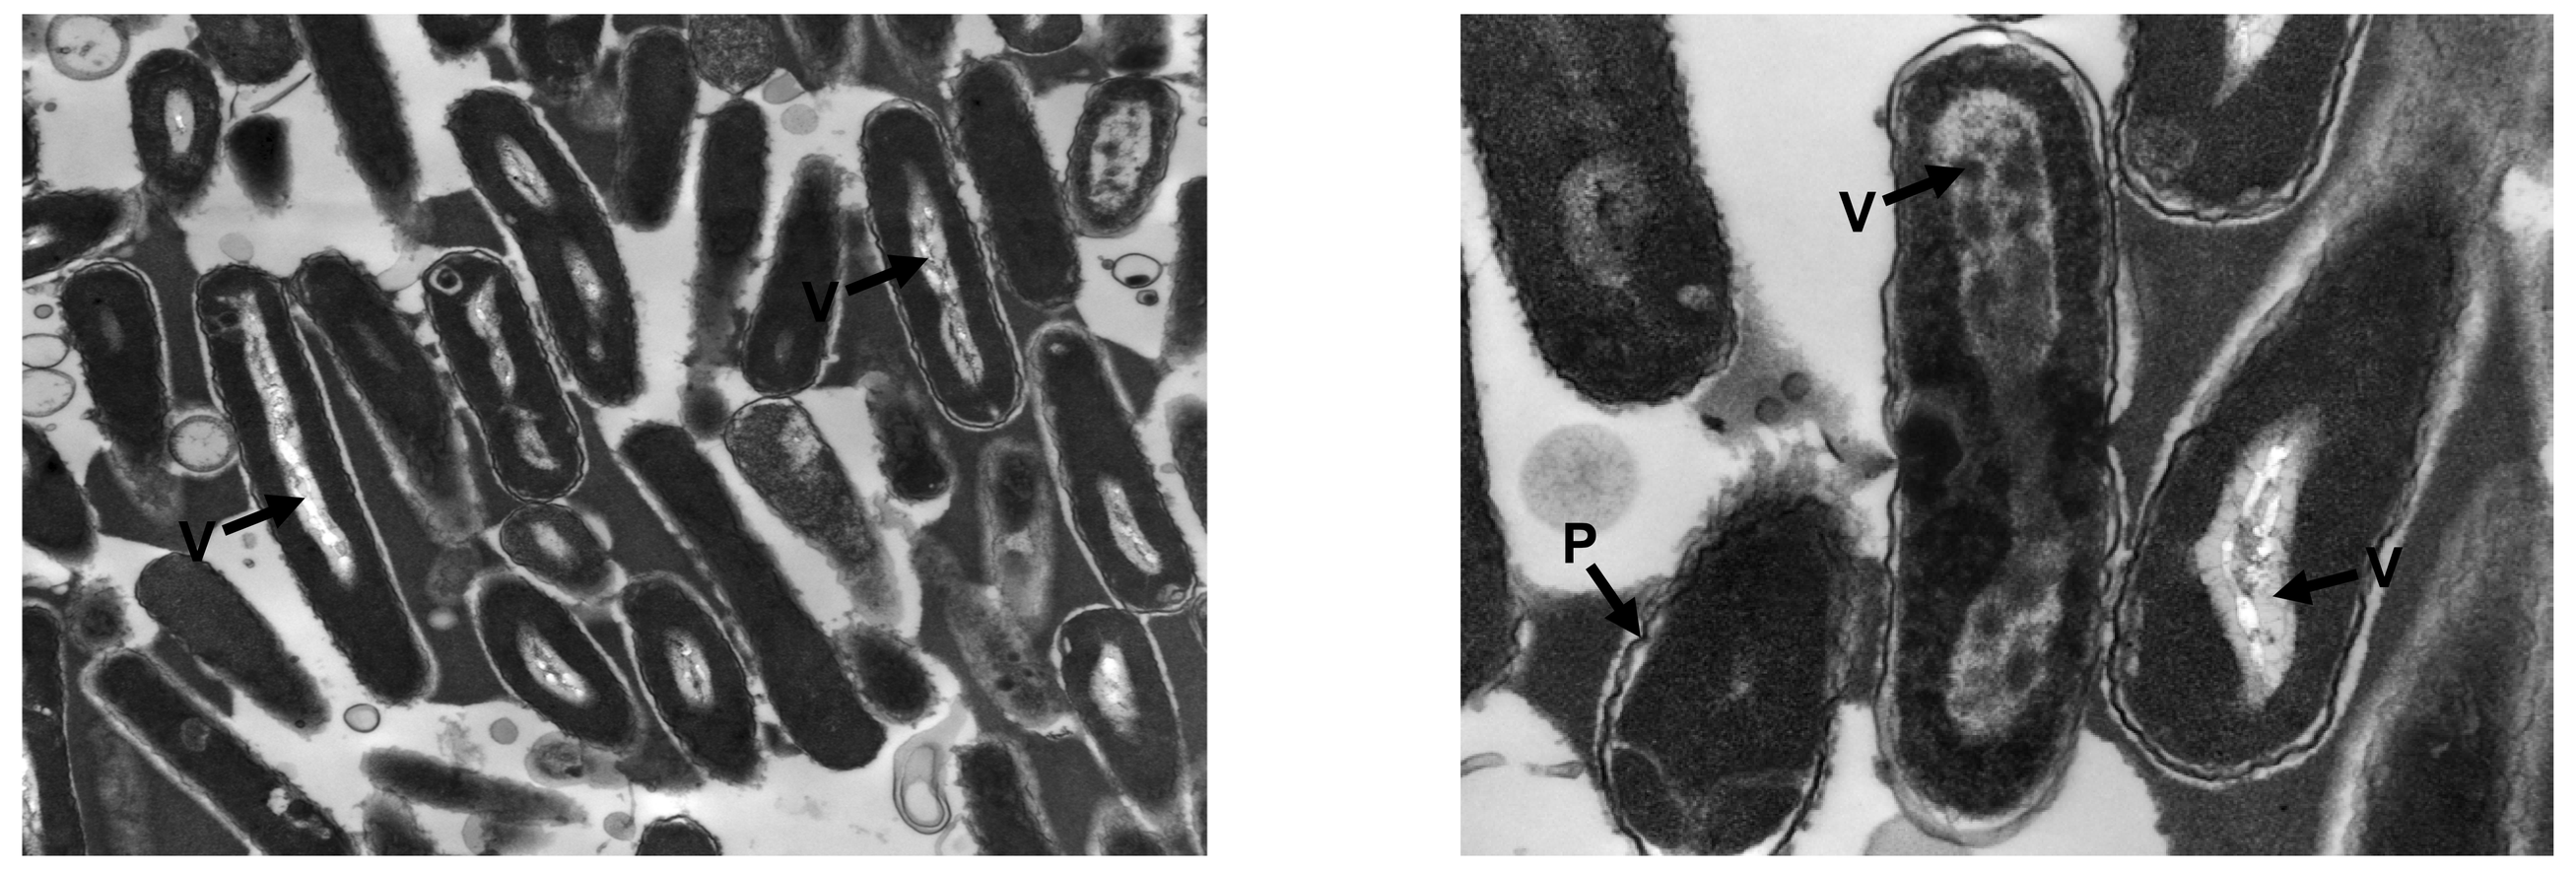
V
V
V
P
V

E-101 causes abnormal elongation, large cytoplasmic vacuoles (V), and pleated cell wall (P).
Left: 9300x, Right: 23000x

METHODS

Bacterial strains. Methicillin-resistant *Staphylococcus aureus* ATCC 43300 and *E. coli* ATCC 25922 were used for time-kill and TEM studies.
Antimicrobial agents. Stock solutions of E-101 enzyme solution pMPO:GO, pMPO alone, GO alone, and substrate solution were prepared at Exoxemis, Inc. (Little Rock, AR). E-101 contains pMPO, GO derived from *Aspergillus niger*, and proprietary amino acids in an aqueous formulation vehicle consisting of 150 mM sodium chloride and 0.02% w/v polysorbate 80 in pH 6.5, 20 mM sodium, phosphate buffer. Solutions containing individual components of pMPO and GO alone were prepared in the same aqueous formulation vehicle. The substrate solution contains 300 mM glucose in the same aqueous formulation as the enzyme solution. The enzyme and substrate solutions are packaged in two separate vials and mixed together to activate the system.
Time-kill assay. The time-kill reaction tubes were prepared to contain the appropriate logarithmic phase growth (10^6 CFU), substrate and enzyme solution (E-101, GO alone, MPO alone). The final concentrations of pMPO and GO in the enzyme solution were 36, 150, and 300 GU/ml and 20, 83, and 167 µg/ml, respectively. Reaction tubes were incubated at 35º C and samples were removed at 0, 5, 15, 30, 60, 90 and 120 minutes for quantitative culture. The log_{10} CFU at each time point was determined.
Transmission electron microscopy (TEM). Bacteria in logarithmic phase growth (109 CFU) were exposed to equivalent volumes of desired enzyme solution with continued incubation at 35º C and sampled at 0, 30, 60 and 120 minutes. Control cultures contained no enzyme solution. Final pellets were fixed in 3% (vol/vol) glutaraldehyde buffered with 0.15M Na cacodylate (pH 7.2), and post fixed in 1% osmium tetroxide. The cells were then dehydrated through an ethanol series and embedded in Spurrs/Polybed epoxy resin. Ultrathin sections were cut and stained with lead citrate and 3% uranyl acetate and examined with a FEI Tecnai G^2 Spirit transmission electron microscope operated at 80 kv. Images were captured with an AMT XR 60 digital camera.

CONCLUSIONS

- Time-kill and TEM studies showed that E-101 oxidative products damage cells in a time- and concentration-dependant manner if pMPO, halide and a source of hydrogen peroxide are present.

- The pMPO-H_2O_2 microbicidal action is several orders of magnitude more potent and than H_2O_2 alone.

- E-101 induced cytoplasmic membrane inclusions (mesosomes) and septal deformation in MRSA. The presence of mesosomes is an indication of an effect on the cytoplasmic membrane.

- E-101 induced cellular elongation (septal deformation), cytoplasmic vacuoles, and pleated cell walls in *E. coli*. These findings are consistent with the multiple mechanisms of E-101 activity.

- The lack of obvious ultrastructure change but a 6 log_{10} kill at 30 minutes of treatment with E-101 suggest that the rapid combustive denaturation of key enzymatic components and/or destruction of membrane integrity are probable microbicidal events.