Microbicidal Activity of the Myeloperoxidase Enzyme System Against Clinically Relevant Bacterial and Fungal Isolates Including Multidrug-Resistant Strains

The Interscience Conference on
Antimicrobial Agents and Chemotherapy, 43rd
ICAAC, Sept. 14-17, 2003
Chicago, IL, USA
Paper # F-1458

S. WOODHEAD[1], G. A. DENYS [2], L. J. HARRELL [3], G. HILL [4], S. VALVANI [4], O. ABRIL-HORPEL [4], W. HAAG [4];
[1]Ricerca Biosciences, Concord, OH, [2]Clarian Health Partners, Inc., Indianapolis, IN, [3]Duke University Medical Center, Durham, NC, [4]ExOxEmis, Inc., Little Rock, AR.

Contact Information
www.exoxemis.com

ABSTRACT

Background: A novel enzyme system utilizing myeloperoxidase (MPO) as a antimicrobial agent has been developed (ExOxEmis, Inc. Little Rock, AR) for topical/local use. The purpose of this study is to investigate the *in vitro* activity of the MPO enzyme system against a broad spectrum of bacterial and fungal isolates implicated in serious infections.
Methods: A total of 196 clinical and reference ATCC isolates were tested including strains with multidrug resistant mechanisms. Isolates included aerobic gram-positive (121) and gram-negative bacteria (64), Candida (5) and Aspergillus (6) species. Late log to early stationary phase cultures were pelleted and resuspended in buffer to ~10^9 cfu/ml. A 1.0 ml volume of enzyme formulation system with organism suspension at a final concentration of ~10^6 to10^7 cfu/ml was tested. The enzyme reaction was then stopped by the addition of catalase. Colony counts were performed and compared to an enzyme diluent control. Time-kill studies were also performed against *S. aureus* ATCC 13709 by measuring the $\log_{10}$ reduction of viable organisms at short exposure times to increasing concentrations of MPO formulation system.
Results: The microbicidal activity of the MPO enzyme system against bacterial isolates occurred within the first 15 min of exposure for 99.0 % of the organisms tested. Complete kill of all bacteria and yeast occurred within 30 min and Aspergillus conidia suspensions within 60 min. Time-kill studies demonstrate that the rate and extent of microbial kill appear to increase with increasing exposure and concentrations of MPO formulation system .
Conclusion: The MPO enzyme system demonstrated rapid and potent cidal activity against a broad spectrum of bacterial and fungal isolates, and may have prophylactic and therapeutic applications.

INTRODUCTION

Myeloperoxidase (MPO) is a major neutrophil protein which is stored in azurophilic granules and released into the phagosome during phagocytosis. The enzyme uses superoxide anion and hydrogen peroxide generated by the neutrophil respiratory burst to form reactive oxidative species that cause cell damage. A cell free oxidant generating enzyme system containing MPO has been developed which exploits this natural antimicrobial system (ExOxEmis, Inc., Little Rock, AR). The mechanism of activity of the MPO enzyme system (Figure 1) involves the use of hydrogen peroxide, generated by a balanced formulation *in-situ*, to convert chloride into hypochlorous acid. Once generated, hypochlorous acid either participates in the direct halogenation of target cell components, or reacts with a second hydrogen peroxide molecule to yield singlet oxygen . Singlet oxygen, a broad spectrum oxidant, is a potent bactericidal agent[1,2]. It exerts its microbicidal action via oxidative destruction of membrane integrity, oxidative inhibition of the enzymes required for metabolic function, and/or oxidative disintegration of nucleic acids required for reproduction. The half-life of singlet oxygen restricts its reactivity to within a 0.2 micron radius at the surface of the targeted bound pathogen. The present study investigated the *in vitro* activity of the MPO enzyme system against a wide range of bacterial and fungal isolates including multi-drug resistant strains.

Figure 1. Myeloperoxidase enzyme system utilizing singlet oxygen.

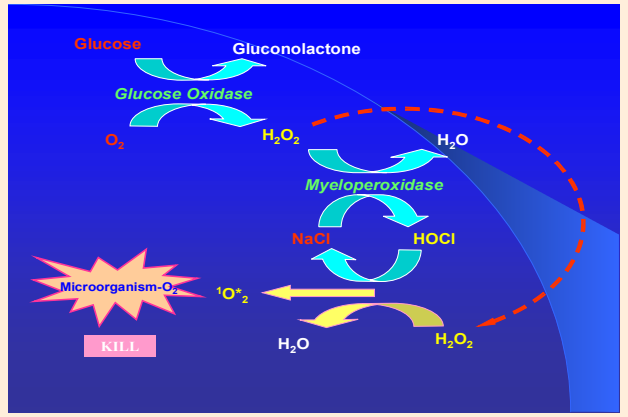
Glucose
Gluconolactone
Glucose Oxidase
O_2
H_2O_2
H_2O
Myeloperoxidase
NaCl
HOCl
Microorganism-O_2
$^1O^*_2$
KILL
H_2O
H_2O_2

Figure 2. *In vitro* activity against enterococci, staphylococci and *S. pneumoniae* strains.

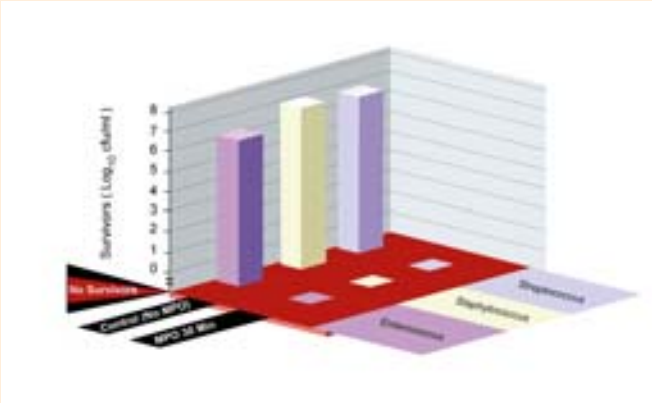

Figure 3. *In vitro* activity against aerobic gram-negative bacteria including multidrug resistant strains.

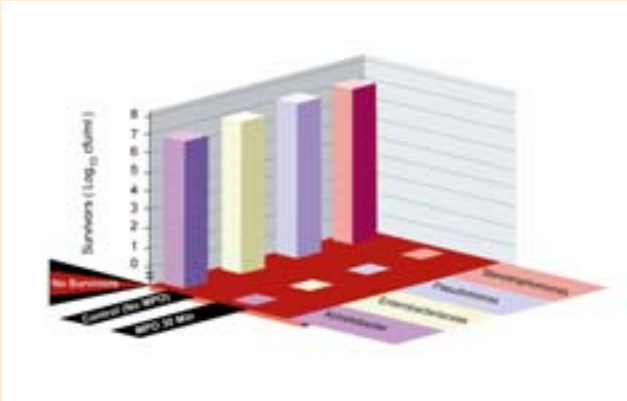

Figure 6. Time-concentration kill curve at short exposure times to MPO formulation against *S. aureus*

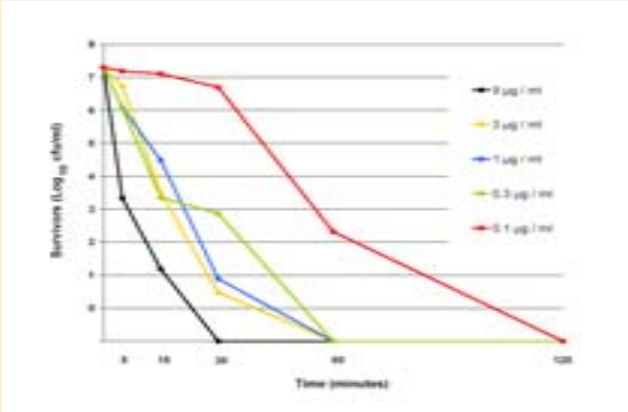
Time (minutes)

Figure 4. *In vitro* activity against selected gram-positive bacterial strains.

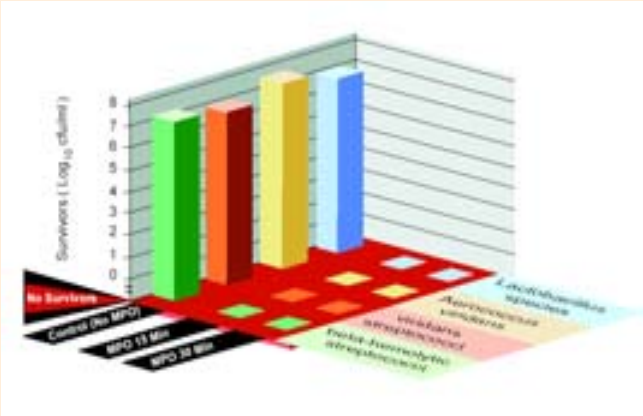
No Survivors
Control (No MPO)
MPO 15 Min
MPO 30 Min

Figure 5. *In vitro* activity against Aspergillus and Candida strains

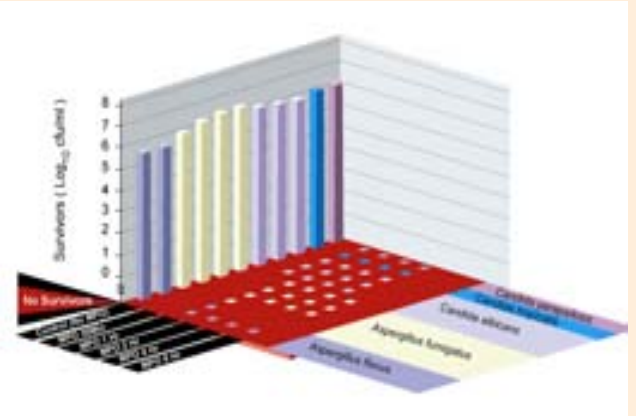

METHODS

Microorganisms: A total of 196 clinical (n=169) and reference ATCC strains (n= 27) comprised of 46 strains of Enterobacteriaceae (including 9 multidrug resistant strains), 13 *Pseudomonas aeruginosa* (including 1 biofilm producing reference strain), 3 *Acinetobacter baumanii*, 2, *Stenotrophomonas maltophilia*, 47 staphylococci (including 10 MRSA, 12 MRCNS, and 1 biofilm producing *S. epidermidis* reference strain), 24 enterococci (including 13 VRE), 21 *S. pneumoniae* (including 10 penicillin non-susceptible strains), 15, beta-hemolytic streptococci, 12 viridans streptococci, 1 *Aerococcus viridans*, 1 Lactobacillus spp, 6 Aspergillus spp., and 5 Candida spp. were tested. These strains were selected to challenge the MPO enzyme system to a broad range of clinically relevant bacterial and fungal strains.

Determination of *In Vitro* Activity: Bacterial and Candida suspensions were prepared by the shake flask method to achieve late log to early stationary phase growth. For fastidious organisms (e.g. *S. pneumoniae*), suspensions were prepared by the direct colony method (stationary phase growth). Aspergillus conidia suspensions were prepared from agar slants by agitation with buffer. All suspensions were than pelleted and resuspended in buffer to ~10^9 cfu/conidia/ml. A 1.0 ml volume of enzyme formulation, containing 6 ug/ml MPO, plus organism suspension at a final target concentration of ~10^6 to 10^7 cfu/ml was tested. Treatment vials were incubated at 37°C in a dry bath. The enzyme reaction was stopped by the addition of catalase (100 ul) at 15 and 30 minutes for bacterial and 30 and 60 minutes for fungal strains. The contents of each vial were inoculated onto isolation media for quantitative culture and incubated for 24 h at 37°C**.** After incubation, colonies were counted and compared to an enzyme diluent control.

Time-kill Studies: The rate and extent of survival of *S. aureus* exposed to low concentrations of MPO containing formulation were determined over time. Culture suspensions were treated as describe above to MPO formulations containing 0.1 to 9 ug/ml MPO and treatment times of 5, 15, 30, 60, and 120 minutes. Quantitative colony counts were performed to determine the time point for maximal kill.

RESULTS

- The *in vitro* activity of the MPO enzyme system against gram positive and gram negative bacteria is presented in Figures 2-4. At 30 minutes or less of treatment with 6 ug/ml of MPO in the formulation, there were no survivors.
- The microbicidal activity of the MPO enzyme system against both antimicrobial susceptible and resistant strains of enterococci, staphylococci and *S. pneumoniae* occurred within 30 minutes (Figure 2). Upon re-testing, there were no survivors within 15 minutes of treatment except for one biofilm producing stain of *S. epidermidis*, which showed no survivors within 30 minutes.
- The MPO enzyme system demonstrated rapid and cidal activity against beta hemolytic and viridan's streptococci (Figure 4). There were no survivors at 15 minutes or less of treatment.
- Among gram negative strains tested, there were no survivors at 15 and 30 minutes of treatment with the MPO enzyme system, regardless of the antimicrobial phenotype of each organism (Figure 3).
- The *in vitro* activity of the MPO enzyme system against Aspergillus and Candida spp. is presented in Figure 5. At 30 minutes for Candida spp. and within 60 minutes for Aspergillus (conidia suspension) spp., of treatment with 6 ug/ml of MPO in the formulation, there were no survivors.
- The results of time-kill studies against *S. aureus* are shown in Figure 6. The data is presented as $\log_{10}$ decrease in survivors relative to the initial inoculum at pre-determined endpoints. The rate and extent of bacterial kill appeared to increase with increasing exposure and concentrations to MPO formulation.

CONCLUSION

- MPO enzyme system is rapidly microbicidal, broad spectrum, and highly active even in the presence of antibiotic resistant strains.
- MPO enzyme system mechanism of action is similar to that of the natural neutrophil host defense system which makes emergence of resistance, as seen with traditional synthetic antimicrobials, potentially unlikely[3].
- MPO enzyme system *in vitro* activity against clinically important pathogens and unique mechanisms of action warrant further investigation for the prevention and treatment of infections.

REFERENCES

(1) Chika, K., Makiuchi, M., Miyazaki, J., Fujinaga, T., and Kakinuma, K. 1999. Physiological production of singlet molecular oxygen in the myeloperoxidase-H202-chloride system. FEBS Letters 443, 154-158.
(2) Tatsuzawa, H., Maruyama, T., Hori, K., Sano, Y., and Nakano, M. 1999. Singlet oxygen as the principal oxidant in myeloperoxidase-mediated bacterial killing in neutrophil phagosome. Biochem. Biophys. Res. Commun. 262, 467-650.
(3) Denys, G.D, Woodhead, S., Becquerelle, S, Valvani, S., Abril-Horpel, O., and Haag, W. 2003. Resistance selection studies with the myeloperoxidase enzyme system against *Staphylococcus aureus*, *Enterococcus faecium*, and *Pseudomonas aeruginosa*. American Society for Microbiology, 43rd Annual ICAAC, Abst. F-353.