DEPARTMENT OF
PATHOLOGY AND LABORATORY MEDICINE
Indiana University School of Medicine

INHIBITORY EFFECT OF WHOLE HUMAN BLOOD ON THE ANTISEPTIC ACTION OF E-101 SOLUTION, A MYELOPEROXIDASE-MEDIATED FORMULATION, COMPARED TO CONVENTIONAL WOUND CLEANSERS

G. A. Denys[1], J.L. Carpenter[1], R.C. Allen[2], and J. T. Stephens, Jr.[3]
[1]Indiana University School of Medicine, Indianapolis, IN, [2]Creighton Univ. Med. Ctr., Omaha, NE, [3]Exoxemis, Inc., Little Rock, AR.

ABSTRACT

Background: E-101 Solution (E-101) is a myeloperoxidase-mediated antimicrobial wound wash for physical irrigation, cleansing, and moisturizing of open wounds. It is composed of porcine myeloperoxidase (pMPO), glucose oxidase (GO), in an aqueous vehicle and activated by the addition of glucose. Once activated, hydrogen peroxide (H_2O_2) is produced in situ by GO dehydrogenation of glucose and reduction of oxygen. The MPO-catalyzed oxidation of chloride ion by H_2O_2 generates hypochlorous acid (HOCl). Once generated, HOCl reacts in a diffusion-controlled reaction with a second H_2O_2 molecule to yield singlet oxygen. We evaluated the effect of blood on the performance of E-101 and three commercially available wound cleansers comprised of stabilized organic derivatives of HOCl.
Materials/methods: Antiseptics were tested against *Escherichia coli* ATCC 8739, *Pseudomonas aeruginosa* ATCC 9027, *Staphylococcus aureus* ATCC 6538, *Candida albicans* ATCC 10231, and *Aspergillus braziliensis* ATCC 16404 in accordance with the antimicrobial effectiveness test USP-51. Comparative antiseptics included Vashe (SteadMed), Microcyn (Oculus Innovative Sciences), and NeutroPhase (NovaBay Pharmaceuticals, Inc.). All antiseptics were tested in the absence and presence of 1, 2, and 5% whole human blood. Time kill studies were also performed with E-101 solution against *P. aeruginosa* ATCC 27853, *S. aureus* ATCC 43300, *E. coli* ATCC 25922, and *Candida auris* CDC B11903 in absence and presence of 2, 5, 10, and 20% blood.
Results: In the USP-51 test, E-101 demonstrated >2-$\log_{10}$ reduction against bacterial and fungal isolates in the presence of 5% blood at day 14 and day 28. With the exception of NeutroPhase vs *S. aureus*, all comparative antiseptics demonstrated <2-$\log_{10}$ reduction in the presence of 5% blood at days 14 and 28. Time-kill results for E-101 against *E. coli* and *P. aeruginosa* showed a >5-$\log_{10}$ reduction in the presence of 2, 5, 10 and 20% blood; for *S. aureus* a >5-$\log_{10}$ reduction in the presence of 2% and 5% blood; for *C. auris* a >5-$\log_{10}$ reduction in the presence of 2% blood.
Conclusions: E-101 remains active in the presence of blood containing catalase and other competitive substances. In contrast, comparative antiseptics with the active component HOCl were easily inactivated by the presence of blood.

INTRODUCTION

E-101 Solution (E-101) is a first-in-class topical myeloperoxidase-mediated formulation developed as an antimicrobial open wound wash solution. It is composed of two enzymes, glucose oxidase (GO) and porcine myeloperoxidase (pMPO) in an aqueous vehicle. Upon topical application of E-101 solution containing glucose, hydrogen peroxide (H_2O_2) is produced in situ by GO that drives pMPO-dependent oxidation of chloride to hypochlorous acid (HOCl). Once generated, HOCl (or its conjugate base OCl- (pKa = 7.5) participates in a diffusion controlled reaction with a second H_2O_2 molecule to yield singlet molecular oxygen ($^1O_2$), a metastable electronically excited reactant with a microsecond lifetime (**Figure 1**). The present study was conducted to measure the performance of E-101 to three predicate antimicrobial solutions in the presence of human blood and blood products.

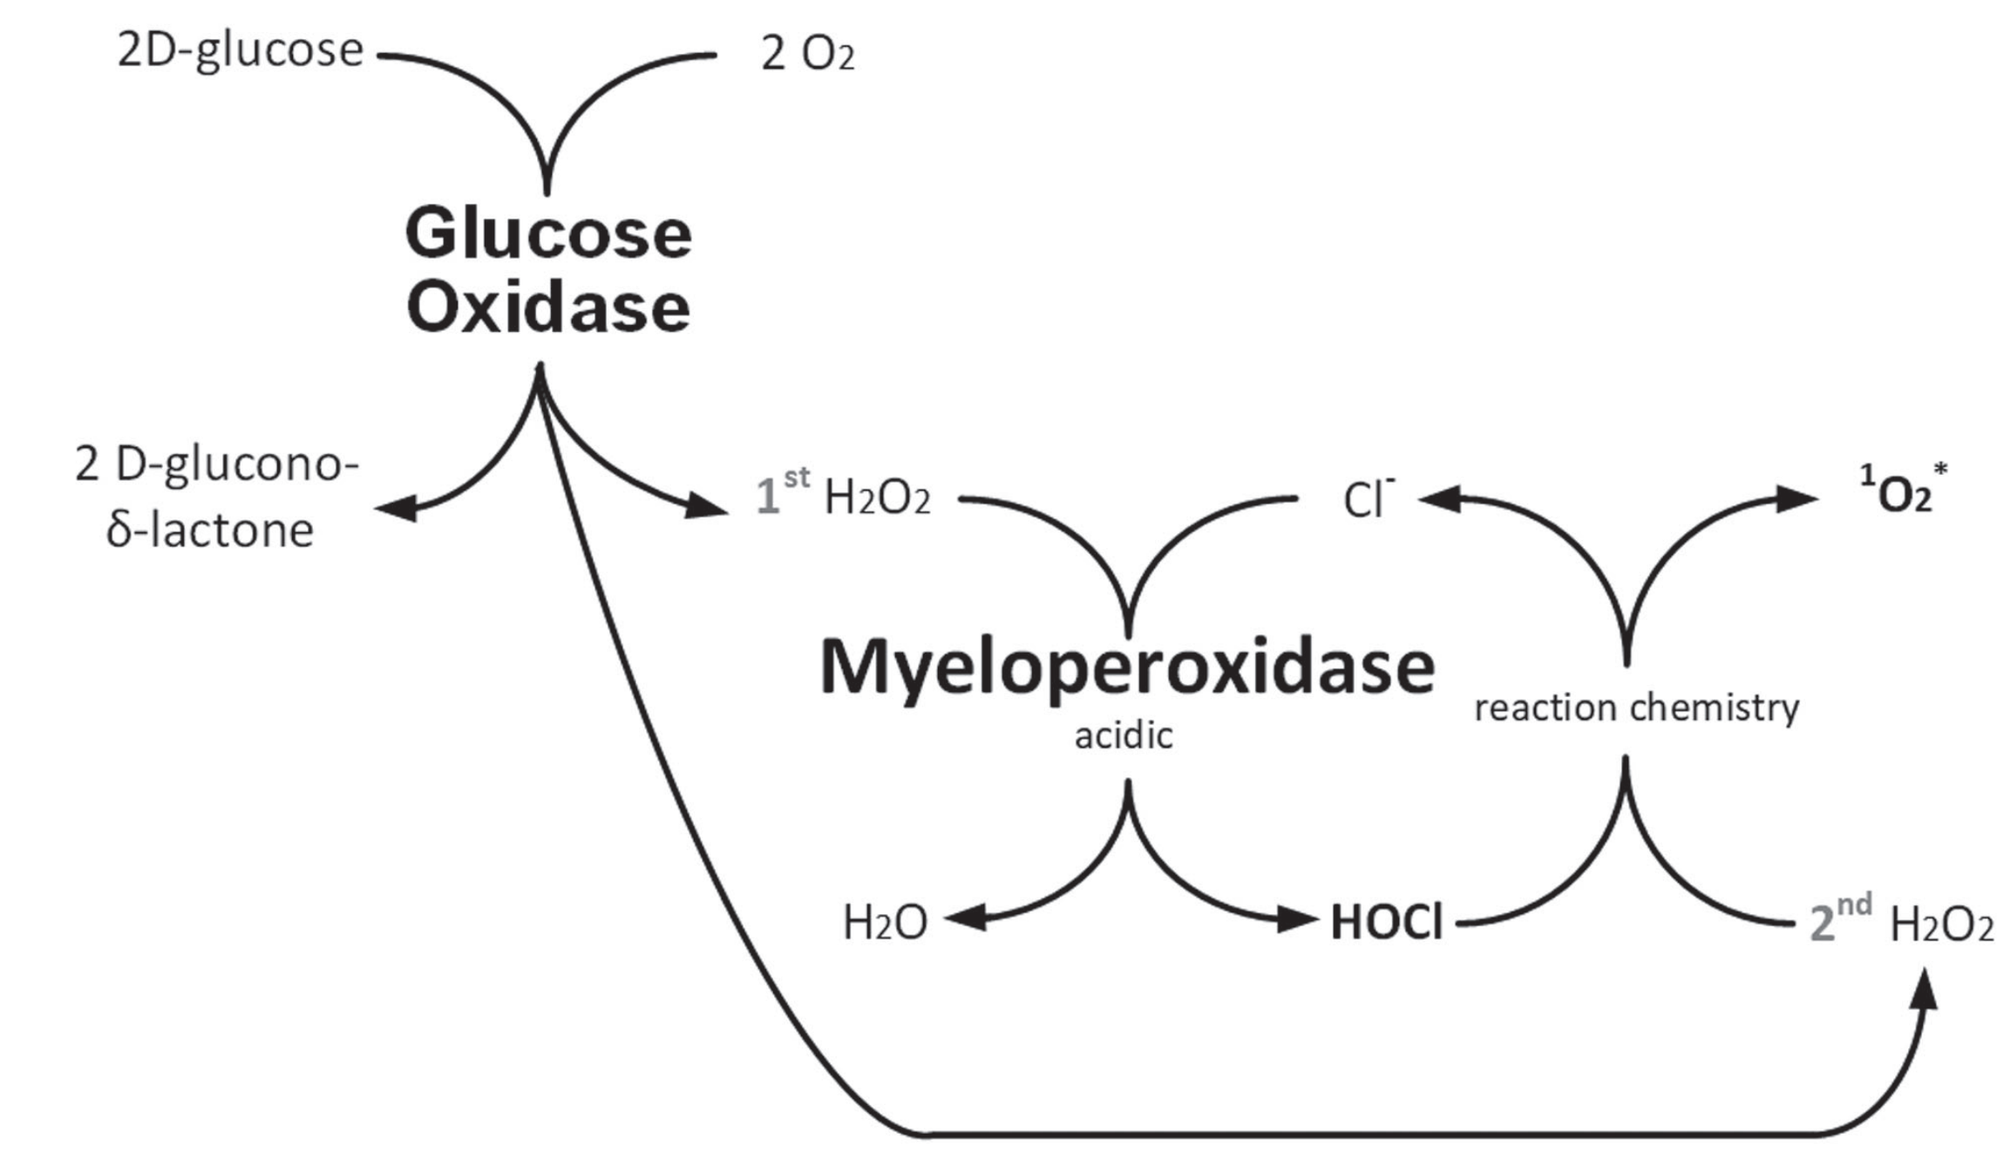
2D-glucose
2 O2
Glucose Oxidase
2 D-glucono-δ-lactone
1st H2O2
Cl-
1O2*
Myeloperoxidase
acidic
reaction chemistry
H2O
HOCl
2nd H2O2

Figure 1. Mechanism of action of E-101 Solution. Singlet oxygen ($^1O_2$*) is a potent electrophilic oxygenating agent capable of reacting with a broad spectrum of electron rich compounds.

RESULTS

Test Product +5%blood	Organism	Log10 Reduction of CFU/mL 1 Day	14 days	28 days
E-101	*C. albicans*	-	2.301	6.447
	S. aureus	2.398	6.301	6.301
	E. coli	6.362	6.362	6.362
	P. aeruginosa	6.462	6.462	6.462
	A.brasiliensis	-	3.331	3.745
NeutroPhase	*C. albicans*	-	-0.456	0.368
	S. aureus	2.382	6.301	4.301
	E. coli	-1.070	0.248	-0.036
	P. aeruginosa	1.064	1.582	0.348
	A.brasiliensis	-	0.854	1.155
Microcyn	*C. albicans*	-	0.049	1.049
	S. aureus	0.155	1.000	-0.591
	E. coli	-0.229	-0.115	-0.036
	P. aeruginosa	-1.880	-1.617	-2.117
	A.brasiliensis	-	1.398	1.398
Vashe	*C. albicans*	-	-0.434	1.067
	S. aureus	0.046	1.561	1.393
	E. coli	-0.195	-0.053	-0.752
	P. aeruginosa	0.781	-1.191	-1.918
	A.brasiliensis	-	1.477	1.247

Table 1. Antimicrobial Effectiveness Test (USP-51). The reactive oxidants of E-101 remains active (>2 Log_{10} reduction from initial inoculum count) in the presence of 5% blood. Comparative antimicrobial wound wash solutions were inactivated in the presence of 5% blood solutions (<2 Log_{10} reduction from initial inoculum count). Only E-101 and NeutroPhase showed activity (>4 Log_{10}) against *S. aureus* in the presence of 5% blood.

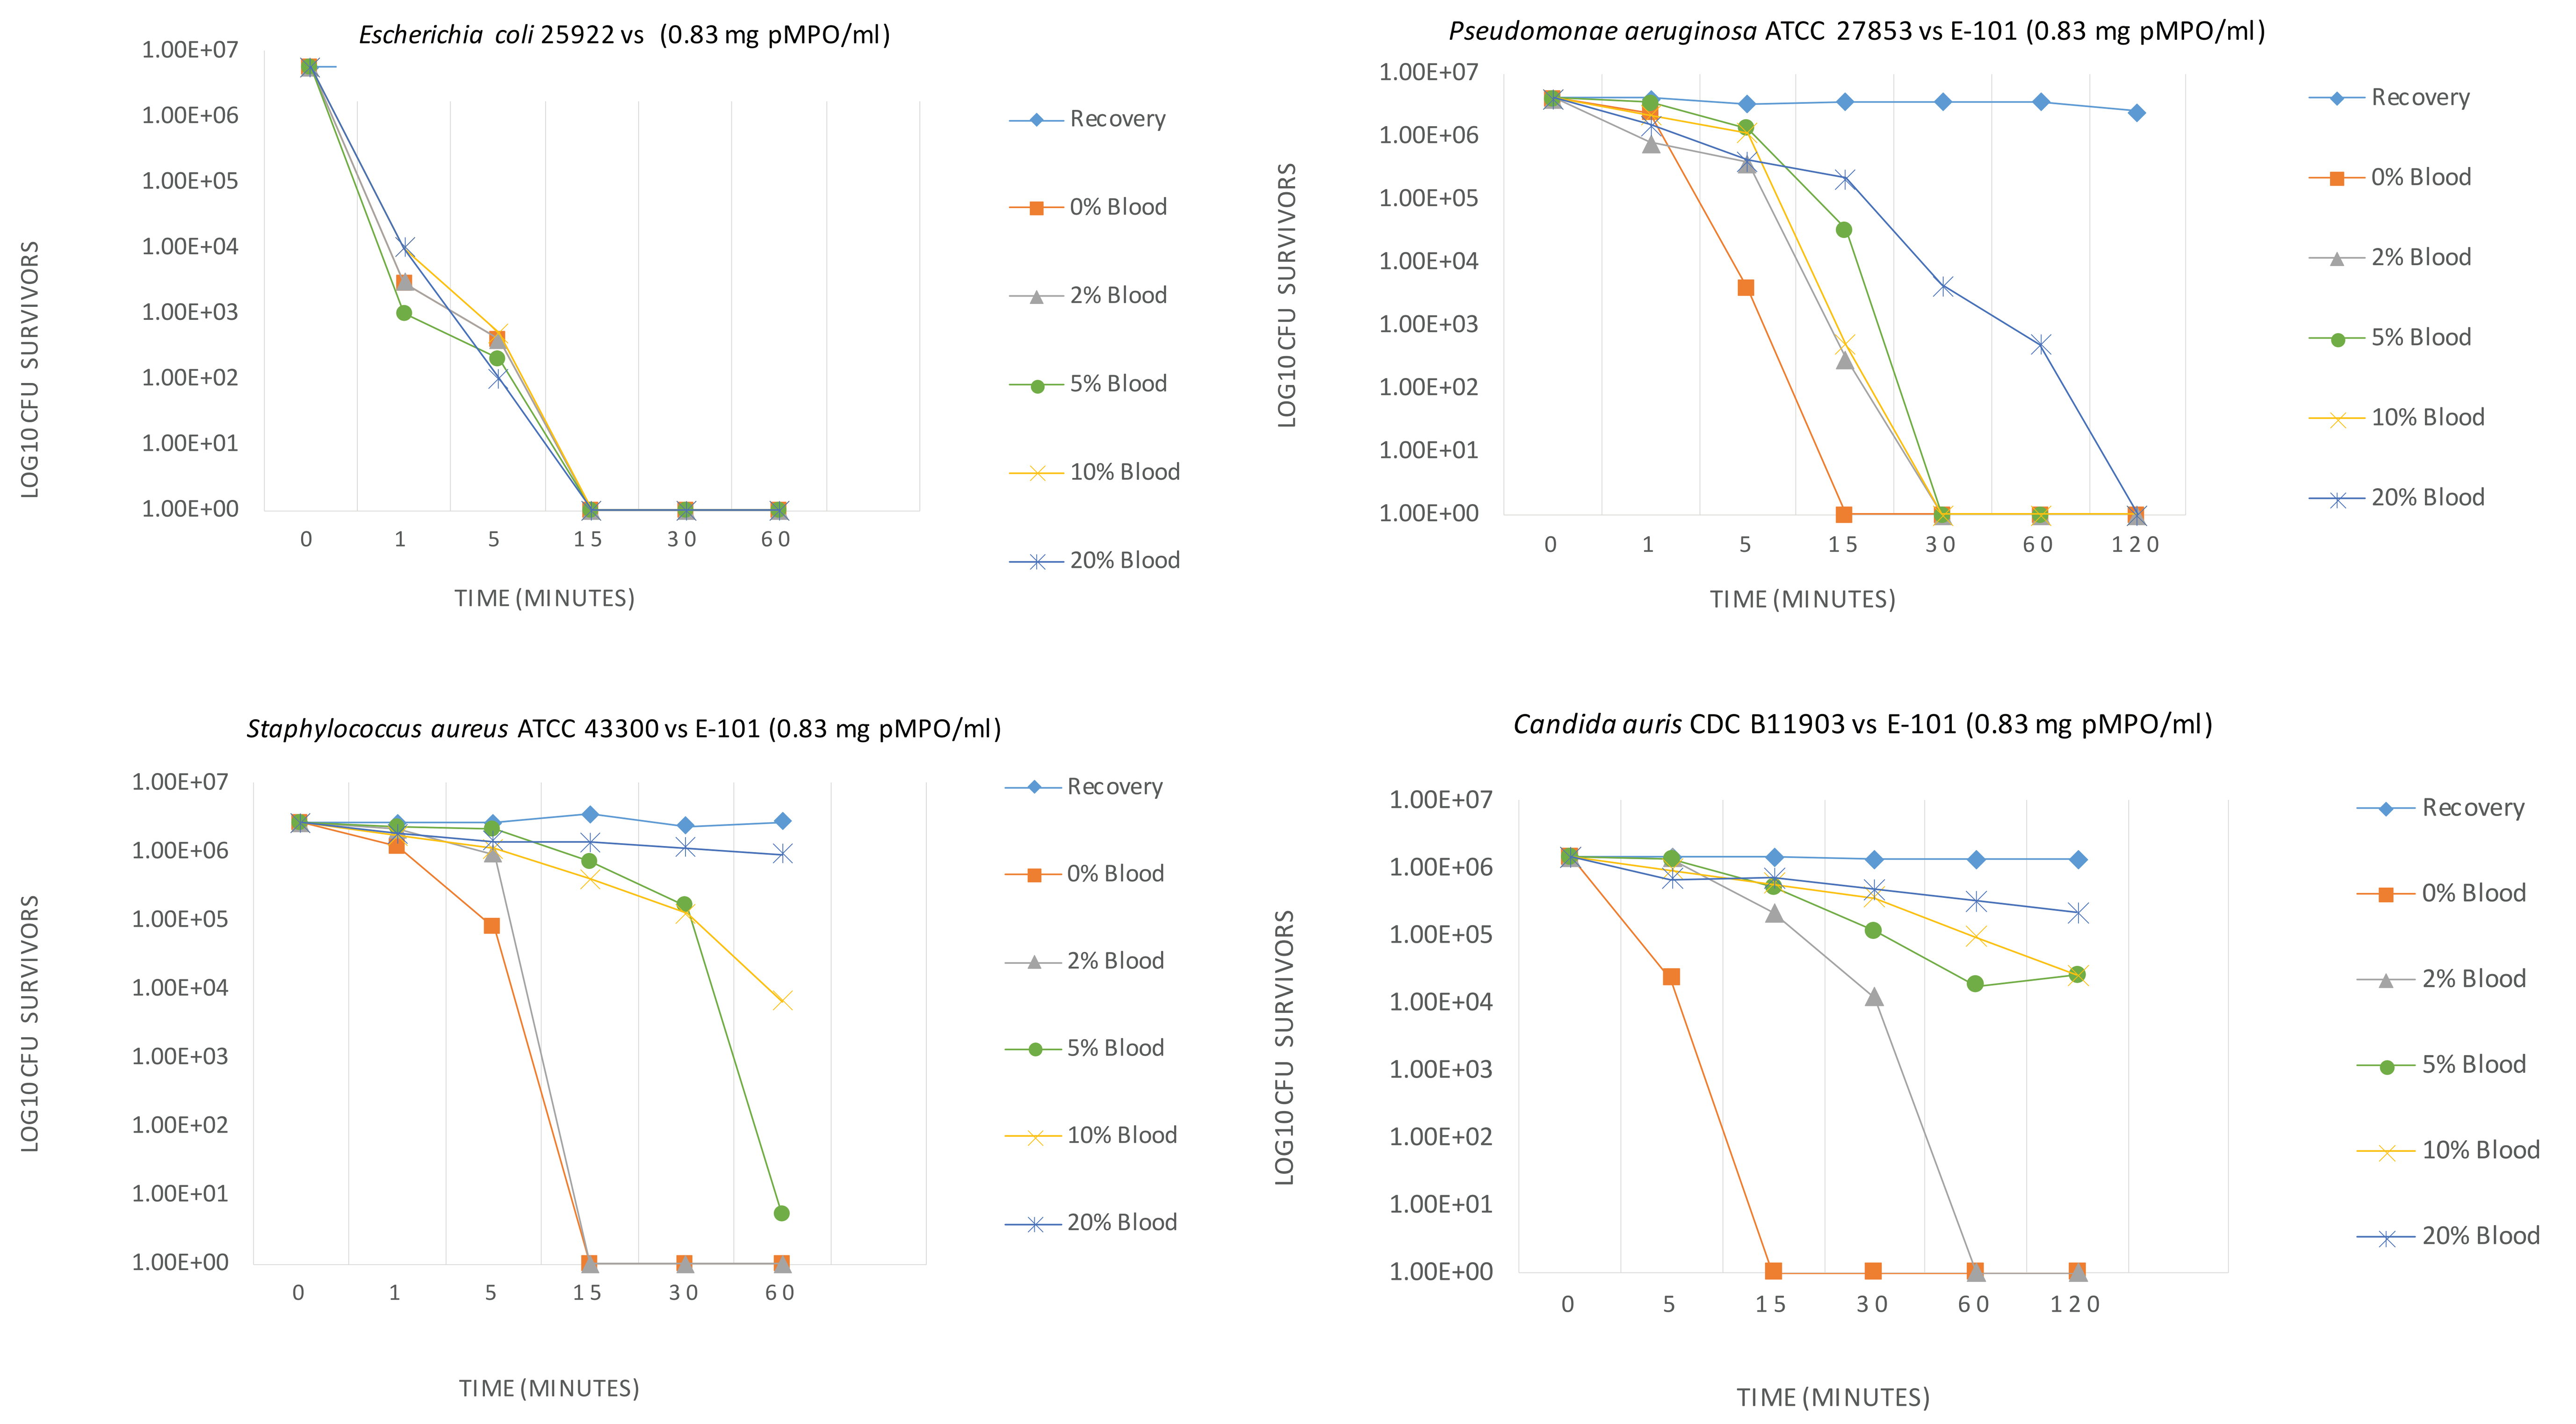
Escherichia coli 25922 vs (0.83 mg pMPO/ml)
Pseudomonae aeruginosa ATCC 27853 vs E-101 (0.83 mg pMPO/ml)
Staphylococcus aureus ATCC 43300 vs E-101 (0.83 mg pMPO/ml)
Candida auris CDC B11903 vs E-101 (0.83 mg pMPO/ml)
LOG10 CFU SURVIVORS
TIME (MINUTES)
Recovery
0% Blood
2% Blood
5% Blood
10% Blood
20% Blood

Figure 2. Time-kill results for E-101 at 0.83 mg pMPO/ml. A bactericidal reduction (>5-$\log_{10}$ viability) of *E. coli* was obtained within 15 min in the presence of 2, 5, 10, and 20% blood. A bactericidal reduction of *P. aeruginosa* was obtained within 30 min in the presence of 2, 5, and 10% blood with continued bactericidal activity in the presence of 20% blood (120 min). A bactericidal reduction of *S. aureus* was obtained within 15 and 60 min in the presence of 2% and 5% blood, respectively. A bactericidal reduction of *C. auris* was obtained within 60 min in the presence of 2% blood.

METHODS

Organisms: USP-51; *Escherichia coli* ATCC 8739, *Pseudomonas aeruginosa* ATCC 9027, *Staphylococcus aureus* ATCC 6538, *Candida albicans* ATCC 10231, and *Aspergillus braziliensis* ATCC 16404. Time-kill; *E. coli* ATCC 25922, *P. aeruginosa* ATCC 27853, *S. aureus* ATCC 43300, and *Candida auris* CDC B11903 7853,
Antiseptics: E-101 Solution, 0.83 mg pMPO/ml (Manufacturer, Exoxemis, Inc., Little Rock, AK), NeutroPhase Skin and Wound Cleaner, 0.03% HOCl (Manufacturer, NovaBay Pharmaceuticals, Inc.), Microcyn Antimicrobial Skin Wound Cleaner, 0.003% HOCl + 0.004% H_2O_2 (Distributor, Oculus Innovative Sciences, Petaluna, CA), and Vashe Wound Therapy+Solution, 0.033% HOCl (Distributor, SteadMed, Fort Worth, TX).
USP-51Test: E-101 and three predicate antimicrobial solutions were tested against five (5) American Type Culture Collection (ATCC) in the presence of 1, 2, and 5% whole human blood. Tests were performed in accordance with the antimicrobial effectiveness test (USP-51)[1] with the modification of added blood. The Log_{10} reduction from the initial population was determined for each test microorganism following exposure of 1, 14, and 28 days post inoculation.

METHODS (CONT)

Time-Kill Assay: Time-kill studies were conducted as previously described[2]. Reaction tubes were prepared to contain log phase growth, activated E-101, and 2, 5, 10 or 20% blood. The final concentration of E-101 was 0.83 mg pMPO/ml. At desired contact times (0, 1, 5 15 30, 60, 120 min), the test mixture was sampled for quantitative culture and incubated at 35oC for 24-48 hrs. The Log_{10} CFU at each time point was determined and compared to growth controls.

CONCLUSIONS

1. E-101 met the passing criteria for the USP-51 Test of >2 Log_{10} reduction from initial inoculum count. The reactive oxidants of E-101 Solution was less susceptible to the inhibitory effect of blood containing catalase and other competitive substrates that competitively react with available $^1O_2$ and hypochlorous acid.

CONCLUSIONS (CONT)

2. Microcyn Antimicrobial Skin Wound Cleanser, Vashe Wound Therapy+Solution, and NeutroPhase Skin and Wound Cleaner all contain a stabilized organic derivative of hypochlorous acid which was inactivated by blood.

3. The time-kill curves demonstrated the rapid bactericidal activity of E-101 in the absence and presence of blood. These data illustrate the superiority of E-101 as an antimicrobial wound wash and skin cleanser.

REFERENCES

1. The United States Pharmacopeial Convention, 2009 USPC Official 8/1/09 – 11/30/09 General Chapters: <51> Antimicrobial Effectiveness Testing. 23 October 2009.
2. Tote K. et al. 2010. Inhibitory effect of biocides on the masses and matrices of Staphylococcus aureus and Pseudomonas aeruginosa biofilms. App. Environ Microbiol 76:3135.